Targeting glutamine metabolism enhances tumor specific immunity by inhibiting the generation and function of suppressive myeloid cells

Min-Hee Oh[1,3], Im-Hong Sun[1], Liang Zhao[1], Im-Meng Sun[1], Wei Xu[1], Chirag Patel[1], Robert Leone[1], Ada Tam[1], Judd Englert[4], Pavel Majer[5], Rana Rais[2], Barbara Slusher[2], Maureen Horton[3], Jonathan D. Powell[1]*

[1]Bloomberg~Kimmel Institute for Cancer Immunotherapy; [2]Johns Hopkins Drug Discovery; [3]Department of Medicine; Johns Hopkins School of Medicine; [4]MedImmune, LLC [5]Institute of Organic Chemistry and Biochemistry, Prague, Czech Republic

Abstract

In order to sustain their inexorable growth, tumors have specialized reprogrammed metabolism. This metabolism creates an acidic, hypoxic and nutrient depleted tumor microenvironment (TME). Such an environment inhibits anti-tumor effector cells while promoting the differentiation and function of inhibitory cells such as T regulatory cells, Myeloid Derived Suppressor Cells (MDSC) and Tumor Associated Macrophages (TAM). We hypothesized that by targeting tumor metabolism we could alter the TME and "condition" tumors to be more susceptible to immunotherapy. To this end, along with the Johns Hopkins Drug Discovery Program we developed a novel prodrug of 6-Diazo-5-oxo-l-norleucine, inhibitor of glutamine metabolism (JHU-083). Recently, it has been shown that M2 macrophages require glutamine metabolism for differentiation and function. In light of the similarities between M2 macrophages and suppressive myeloid cells, we hypothesized that JHU-083 might inhibit the generation and function of MDSCs and TAMs. We tested this hypothesis in the 4T1 breast cancer model and 3LL lung carcinoma model. These tumors are relatively resistant to immunotherapy and are characterized by increased generation of MDSCs and distant spontaneous metastasis. JHU-083 treated mice suppressed tumor growth compared to the vehicle treated group. Immunologically, we observed markedly reduced numbers of MDSCs in circulating blood within 3 days of drug treatment compared to vehicle group, leading to favorable CD8 to MDSCs ratios. Consistently, JHU083 treated group displayed significantly decreased percentages and numbers of MDSCs in the tumor, and increased tumor infiltrating CD8 cells. Interestingly, JHU-083 treatment induced TAM reprogramming. While the TAM from the vehicle treated group displayed increased M2 markers and arginase, the JHU-083 treated tumor infiltrating cells showed increased TNF-a producing M1-like macrophage phenotypes compared to vehicle group. These TNF-a producing cells were negatively correlated with tumor sizes. Notably, JHU083 treatment not only controlled primary tumor growth but also drastically reduced spontaneous lung metastasis. The decrease of MDSCs infiltration in the lung were also observed in JHU083 treated group. Mechanistically, our data suggests that JHU-083 inhibits CSF2/CSF3 production and survival of the tumor itself as well as directly affects macrophage metabolism and signaling. Also, LC-MS based metabolites analysis from JHU-083 treated tumors revealed reduced Kynurenine:tryptophan ratios compared to the control group, indicating the metabolic modulation of the tumor microenvironment. Overall, our data support a novel role for glutamine inhibitor, JHU-083 in enhancing tumor specific Immunity by targeting suppressive myeloid cells.

Introduction

- **Increased monocyte derived suppressive cells are responsible for poor responses to immunotherapy and spontaneous metastasis in 4T1 (mammary) tumors**
- **M2 macrophages rely on glutamine metabolism for differentiation and its function**
- **Monocyte derived suppressive cells (MDSCs) share metabolic characteristics and markers with M2 macrophages**
- **Together with the Johns Hopkins Drug Discovery Program, we developed a novel prodrug of 6-Diazo-5-oxo-l-norleucine, to inhibit glutamine metabolism (JHU-083).**

Objectives

We hypothesized that glutamine antagonism might enhance immunotherapy by regulation of the generation and functions of the suppressive myeloid cells

Results

4T1 tumor is resistant to immune checkpoint blockade immunotherapy

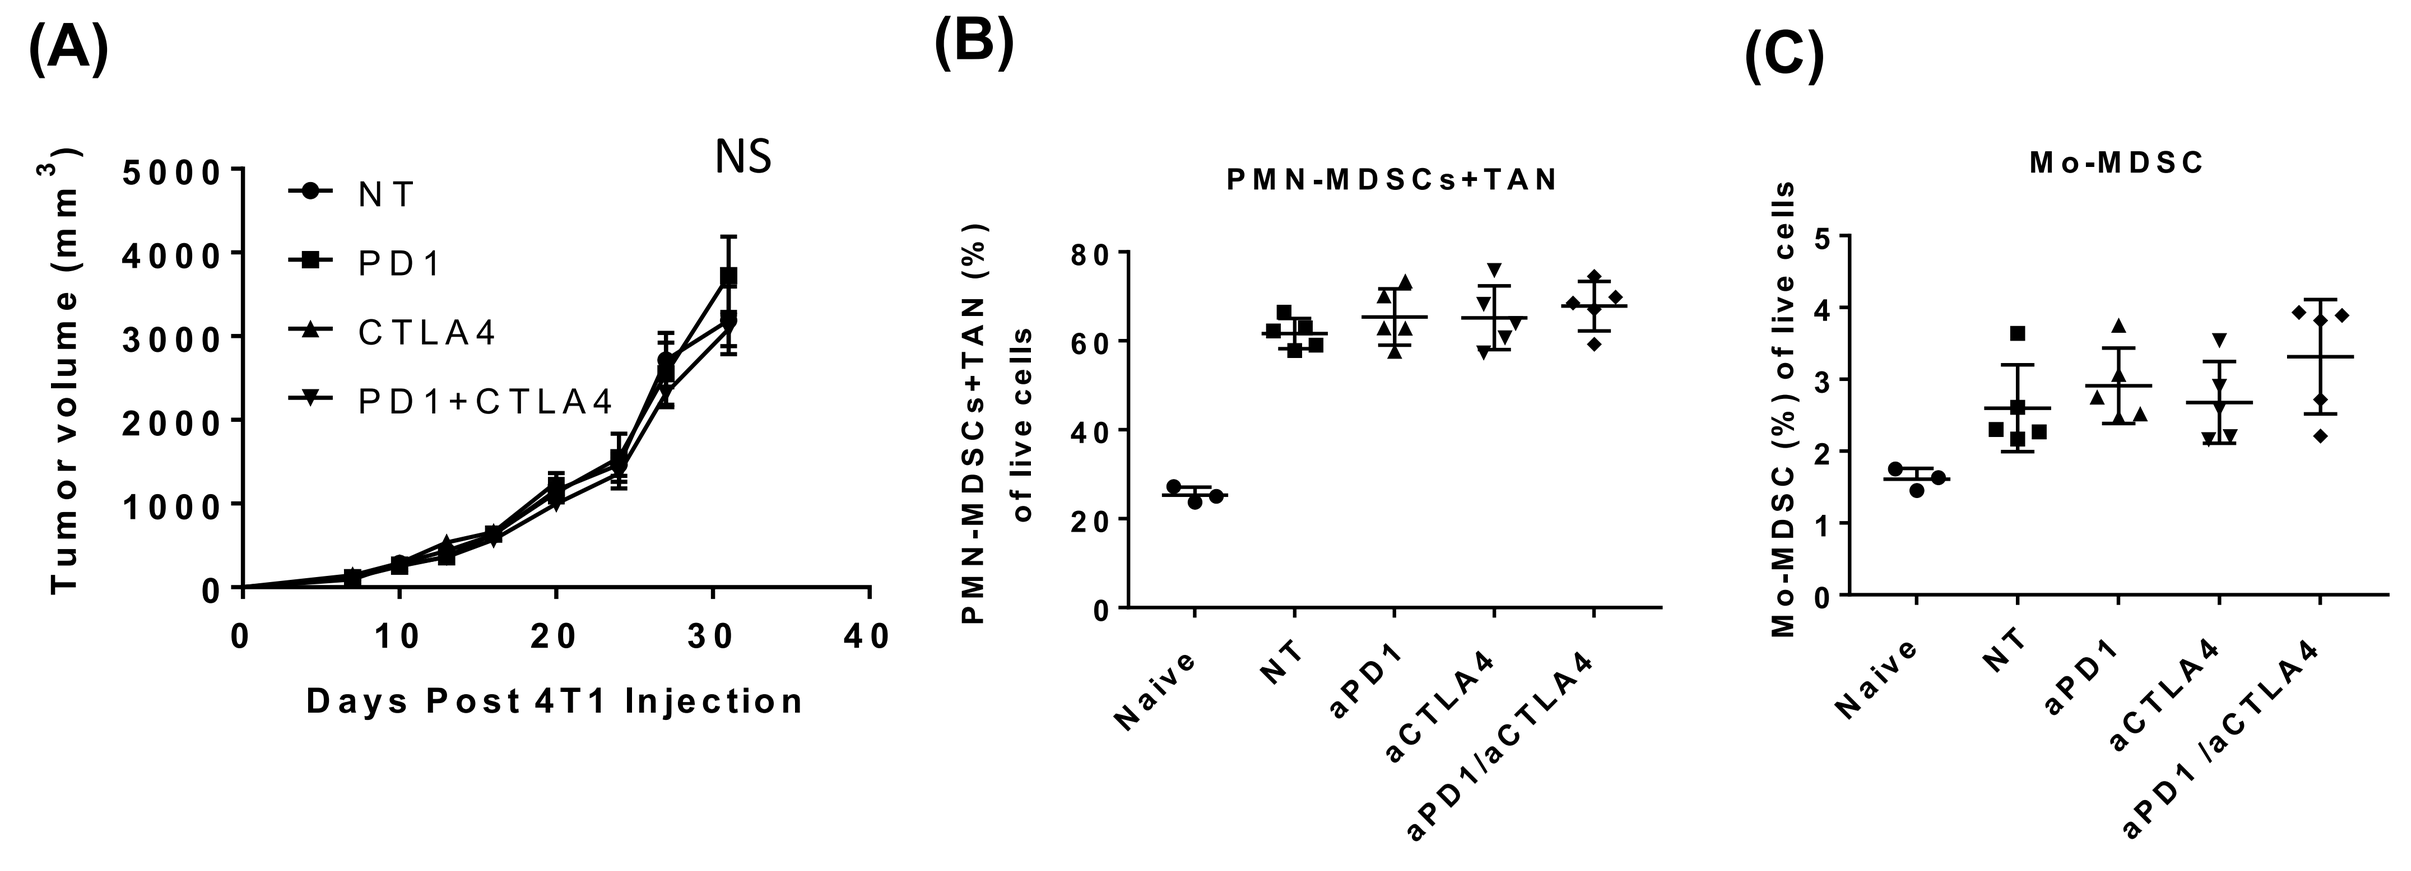
(A)
NS
Tumor volume (mm³)
NT
PD1
CTLA4
PD1+CTLA4
Days Post 4T1 Injection
(B)
PMN-MDSCs+TAN
PMN-MDSCs+TAN (%) of live cells
Naive
NT
aPD1
aCTLA4
aPD1/aCTLA4
(C)
Mo-MDSC
Mo-MDSC (%) of live cells
Naive
NT
aPD1
aCTLA4
aPD1 /aCTLA4

Fig 1. (A) 4T1 tumor is resistant to immune checkpoint blockade (B and C) Immune checkpoint blockade doe not alter increased MDSCs in the blood from 4T1 tumor bearing mice (PMN-MDSC and TAN (polymorphonuclear-MDSCs and tumor-associated neutrophils : CD11b+F4/80-Ly6CloLy6Ghi) and Mo-MDSC (monocytic MDSC : CD11b+F4/80-Ly6ChiLy6Gneg)

Results

Targeting glutamine using JHU-083 inhibits growth of 4T1 tumor and leads to decreased MDSCs

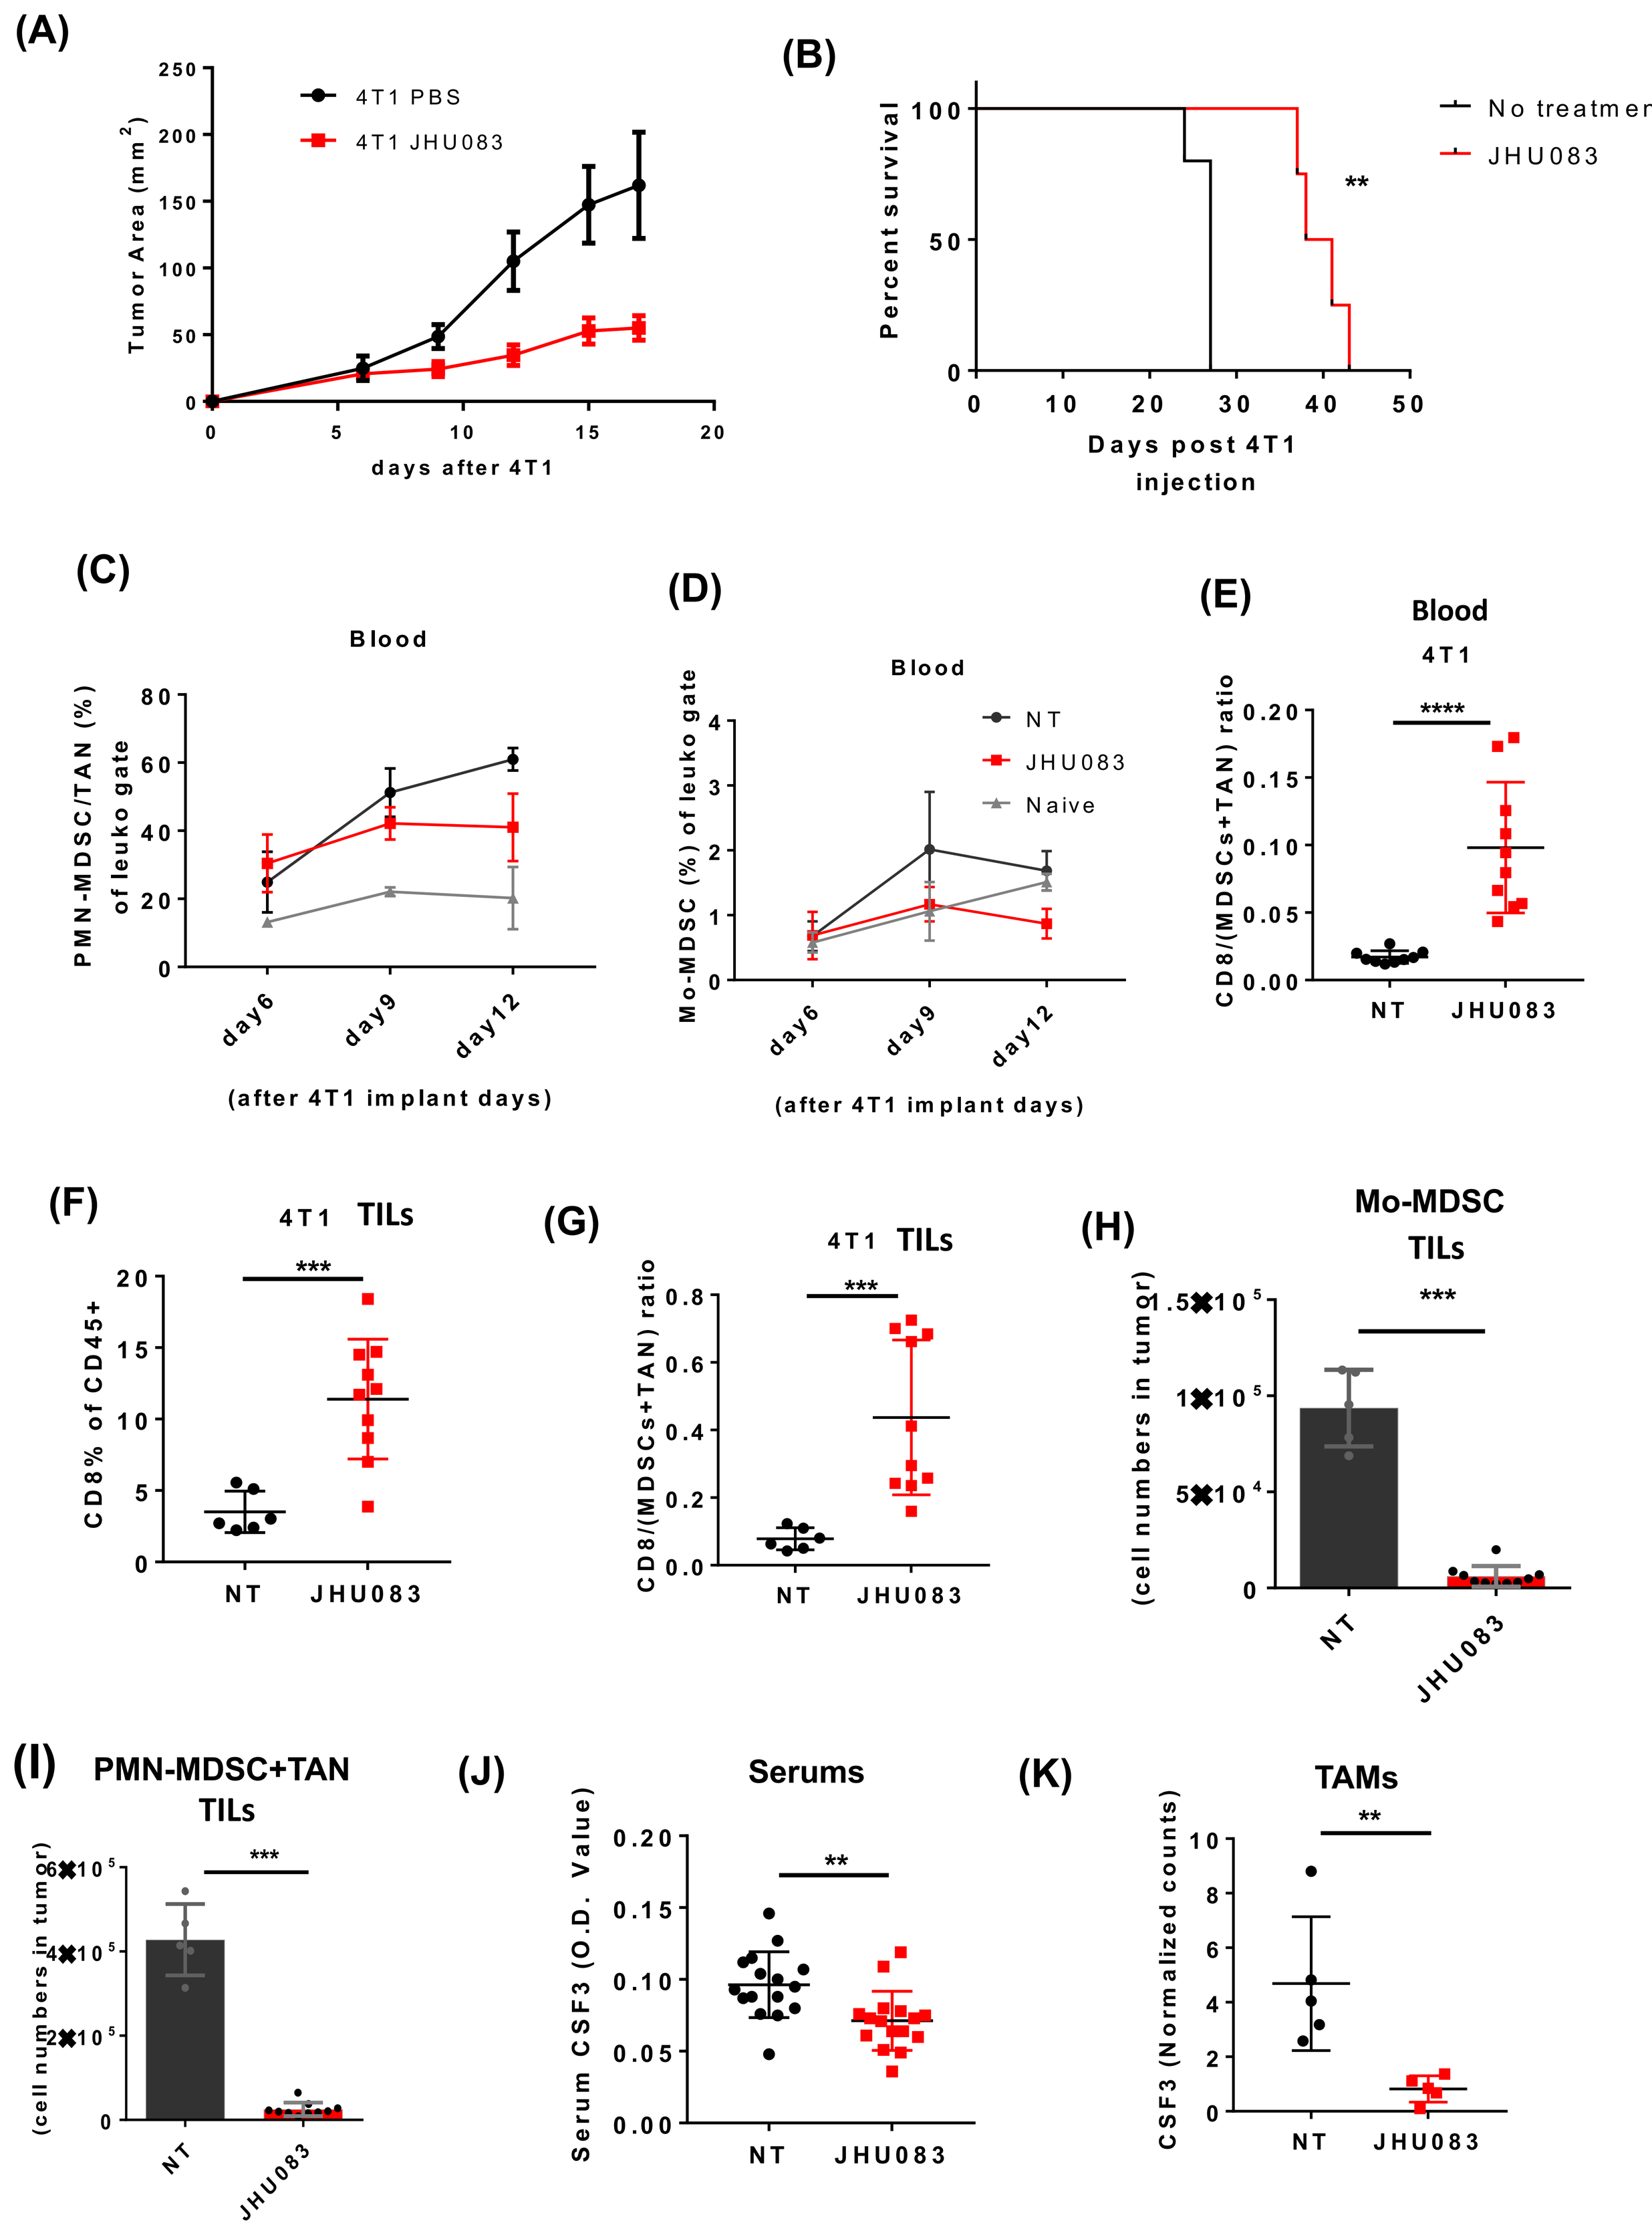
(A)
4T1 PBS
4T1 JHU083
Tumor Area (mm²)
days after 4T1
(B)
No treatment
JHU083
Percent survival
Days post 4T1 injection
(C)
Blood
PMN-MDSC/TAN (%) of leuko gate
day6
day9
day12
(after 4T1 implant days)
(D)
Blood
NT
JHU083
Naive
Mo-MDSC (%) of leuko gate
(after 4T1 implant days)
(E)
Blood
4T1
CD8/(MDSCs+TAN) ratio
NT
JHU083
(F)
4T1 TILs
CD8% of CD45+
(G)
4T1 TILs
CD8/(MDSCs+TAN) ratio
(H)
Mo-MDSC
TILs
(cell numbers in tumor)
(I)
PMN-MDSC+TAN
TILs
(cell numbers in tumor)
(J)
Serums
Serum CSF3 (O.D. Value)
(K)
TAMs
CSF3 (Normalized counts)

Fig 2. Targeting glutamine using JHU-083 in the 4T1 tumor bearing mice showed (A) delayed 4T1 tumor growth (B) increases survival (C) decreased % of PMN-MDSCs and TANs and (D) Mo-MDSCs in blood (E) increased ratio of CD8+ to MDSCs and TANs in the blood (F) increased % of CD8+ in TIL (tumor-infiltrating leukocytes) (G) increased ratio of CD8+ to MDSCs and TANs in TIL (H) reduced the relative numbers of Mo-MDSCs and (I) PMN-MDSCs and TANs (J) decreased CSF3 in the serum (K) decreased CSF3 mRNA in TAMs

Targeting glutamine using JHU-083 markedly reduces kynurenine

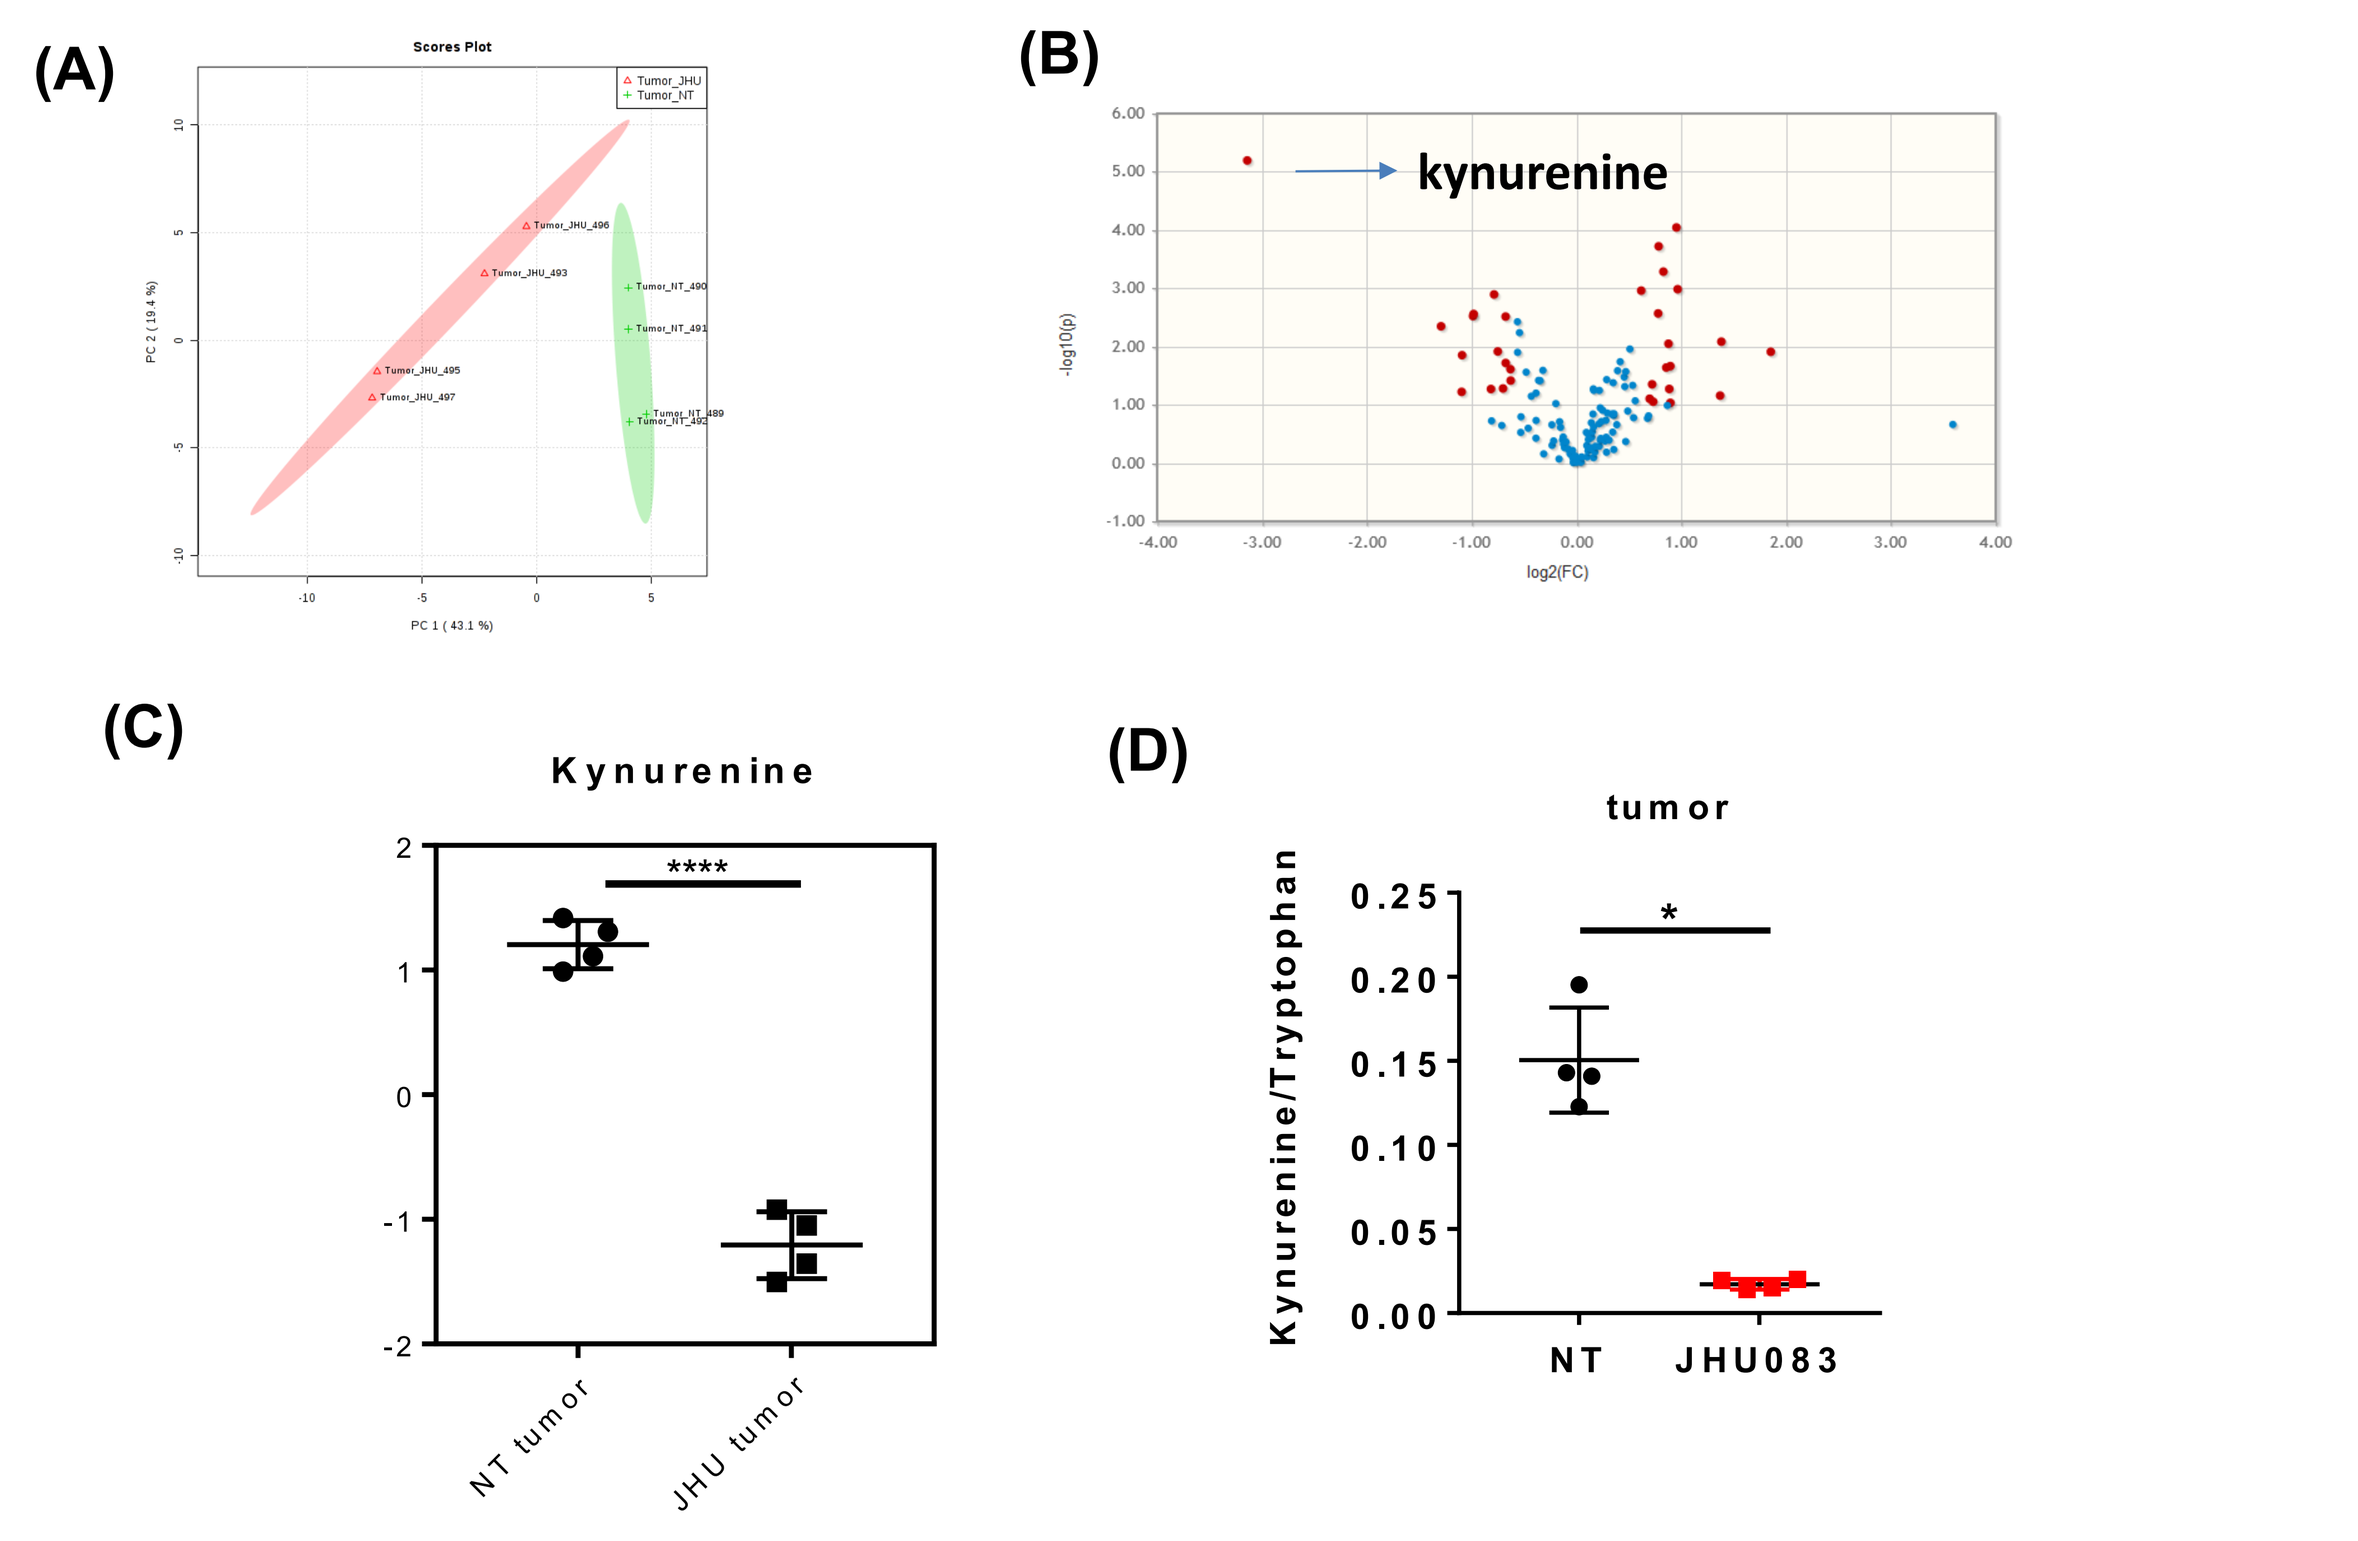
(A)
Scores Plot
(B)
kynurenine
log2(FC)
(C)
Kynurenine
NT tumor
JHU tumor
(D)
tumor
Kynurenine/Tryptophan
NT
JHU083

Fig 3. LC-Mass spec of metabolites of tumors from the NT and glutamine antagonist JHU-083 groups revealed (A) two distinct metabolic clusters correlated between experimental groups (B and C) markedly reduced kynurenine in volcano plot of metabolites (D) decreased ratio of kynurenine to tryptophan

Targeting glutamine increases pro-inflammatory TAMs

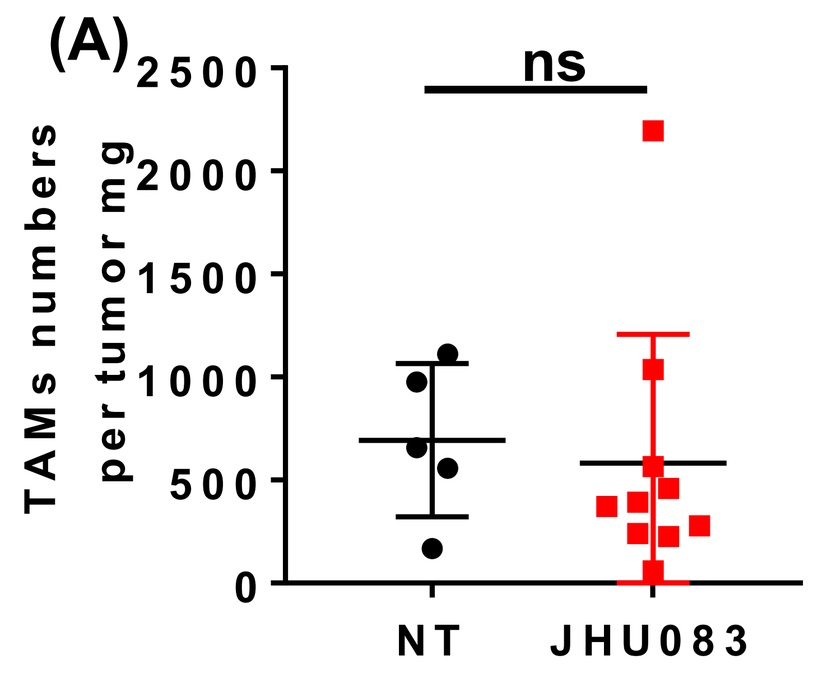
(A)
ns
TAMs numbers per tumor mg
NT
JHU083

(B)

GO molecular function analysis in up-regulated genes in JHU treated TAMs

Term	Count	%	P-Value	Fold Enrichment
Lysosome	39	0.2	2.9E-14	4
Toll-like receptor signaling pathway	22	0.1	0.000031	2.7
Sphingolipid metabolism	12	0.1	0.00037	3.5
Endocytosis	32	0.2	0.00034	2
Hematopoietic cell lineage	18	0.1	0.00032	2.6
Chemokine signaling pathway	28	0.2	0.0014	1.9
Circadian rhythm	6	0	0.0025	5.7
Complement and coagulation cascades	15	0.1	0.0024	2.5
MAPK signaling pathway	36	0.2	0.0023	1.7
Renal cell carcinoma	14	0.1	0.0035	2.5
Cytokine-cytokine receptor interaction	33	0.2	0.004	1.7
Focal adhesion	27	0.2	0.0088	1.7
GnRH signaling pathway	16	0.1	0.011	2
TGF-beta signaling pathway	14	0.1	0.022	2
Fructose and mannose metabolism	8	0	0.027	2.7
Type II diabetes mellitus	9	0.1	0.041	2.3

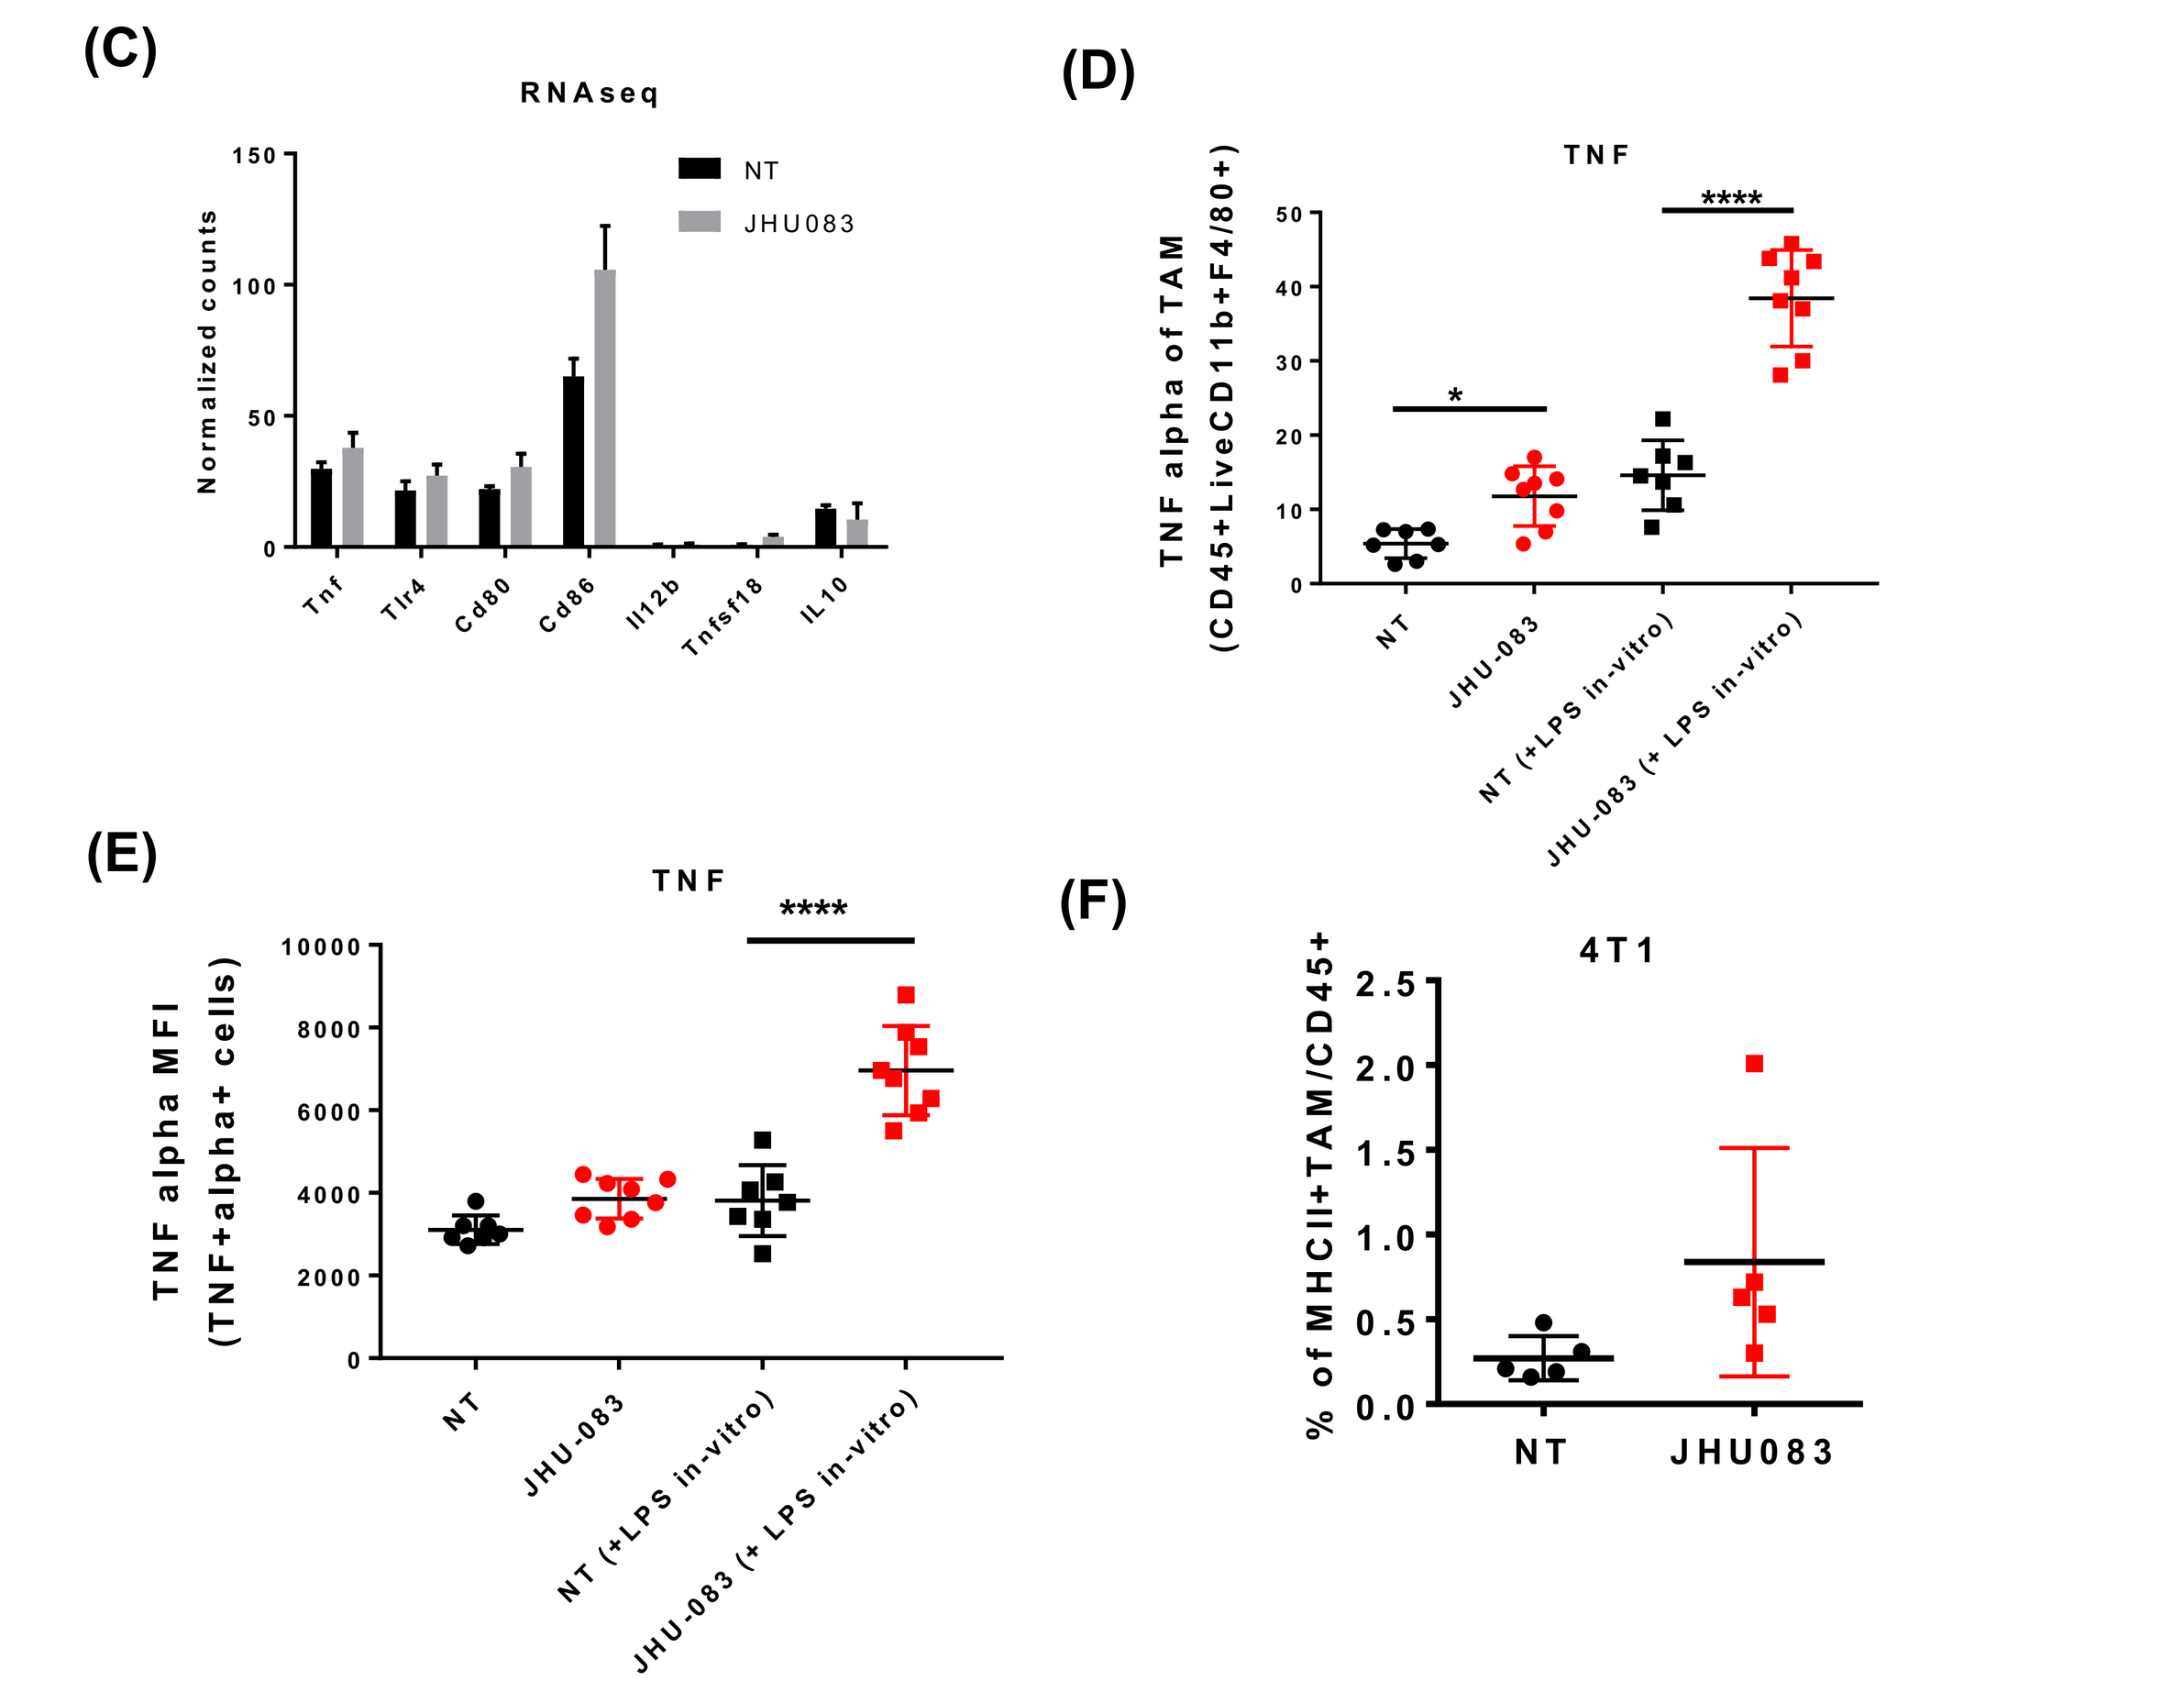
(C)
RNAseq
NT
JHU083
Normalized counts
Tnf
Tlr4
Cd80
Cd86
Il12b
Tnfsf18
IL10
(D)
TNF
TNF alpha of TAM (CD45+LiveCD11b+F4/80+)
NT
JHU-083
NT (+LPS in-vitro)
JHU-083 (+ LPS in-vitro)
(E)
TNF
TNF alpha MFI (TNF+alpha+ cells)
(F)
4T1
% of MHCII+TAM/CD45+
NT
JHU083

Fig 4. (A) Targeting glutamine using JHU-083 in the 4T1 tumor does not alter the numbers of tumor-associated macrophages (TAMs) (B and C) Increased gene expressions related to lysosome and TLR signaling in TAMs (D) increased % of TNF+ TAMs (E) increased amounts of TNF secretion in TAMs (E) increased MHCII+ TAMs from the glutamine antagonist JHU-083 treated mice

Glutamine antagonist JHU-083 inhibits spontaneous lung metastasis

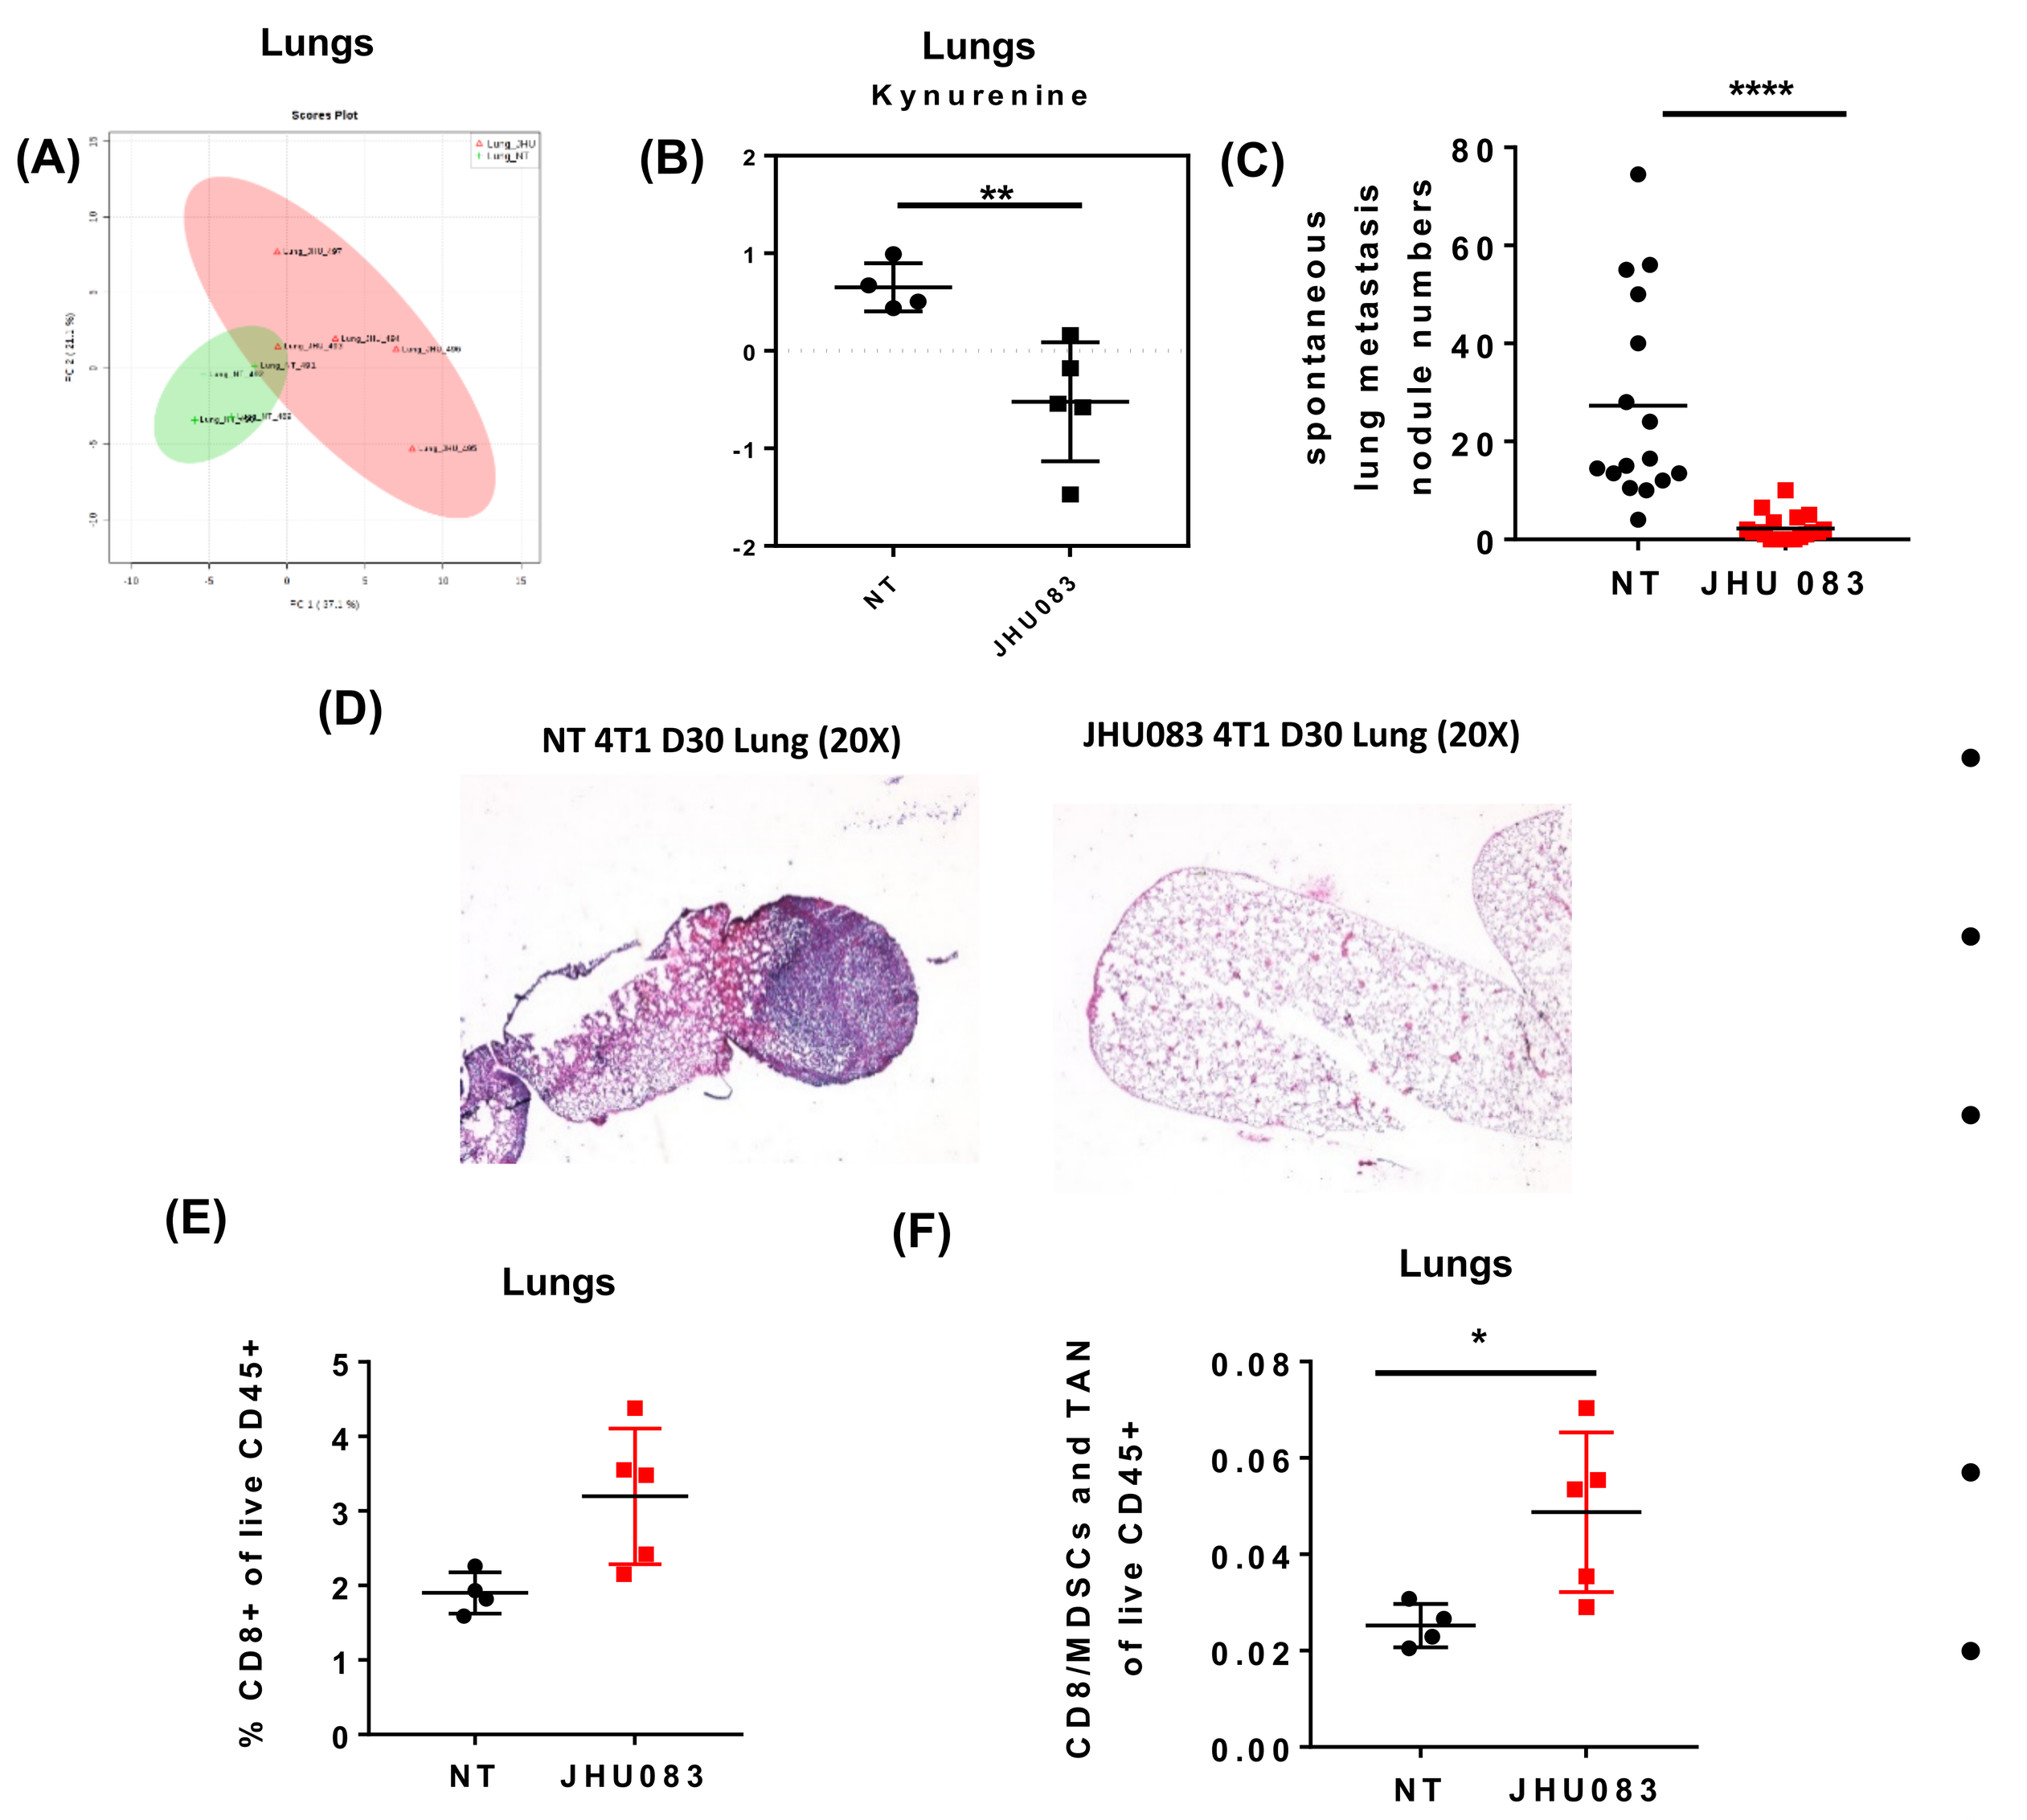
Lungs
(A)
Scores Plot
Lungs
Kynurenine
(B)
NT
JHU083
(C)
spontaneous lung metastasis nodule numbers
NT
JHU 083
(D)
NT 4T1 D30 Lung (20X)
JHU083 4T1 D30 Lung (20X)
(E)
Lungs
% CD8+ of live CD45+
(F)
Lungs
CD8/MDSCs and TAN of live CD45+

Fig 5. (A) LC-Mass spec of metabolites of spontaneous metastatic site (lung) from the NT and glutamine antagonist JHU-083 groups revealed two distinct metabolic clusters (B) markedly reduced kynurenine in volcano plot of metabolites. (C and D) Reduced spontaneous lung metastasis (E) increased % of CD8+ in TIL (tumor-infiltrating leukocytes) (G) increased ratio of CD8+ to MDSCs and TANs in cells from lung of 4T1 tumor bearing mice

Glutamine antagonism enhances priming of tumor antigen

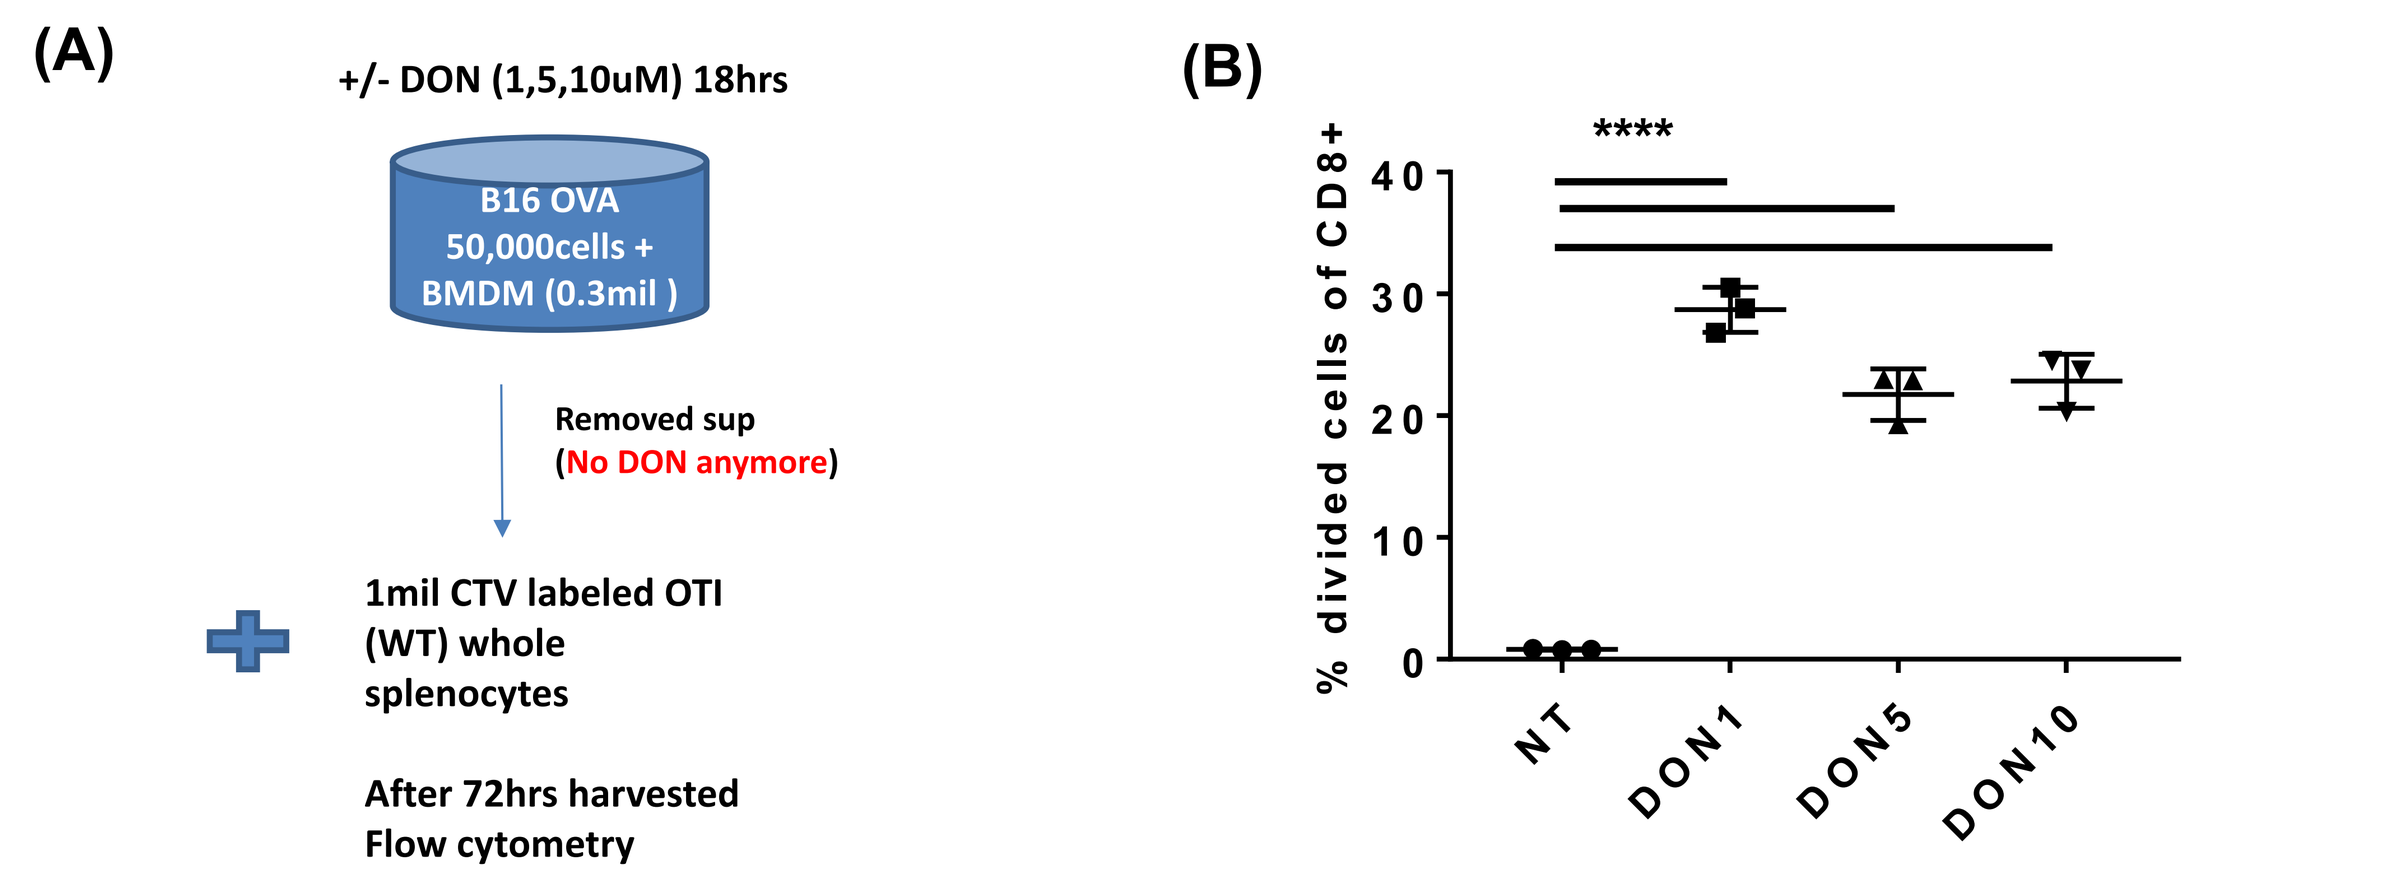
(A)
+/- DON (1,5,10uM) 18hrs
B16 OVA
50,000cells +
BMDM (0.3mil)
Removed sup
(No DON anymore)
1mil CTV labeled OTI
(WT) whole
splenocytes
After 72hrs harvested
Flow cytometry
(B)
% divided cells of CD8+
NT
DON1
DON5
DON10

Fig 6. (A) Scheme of T cell priming experiment (B) Increased tumor antigen specific T cell priming

Glutamine inhibition enhances immunotherapy

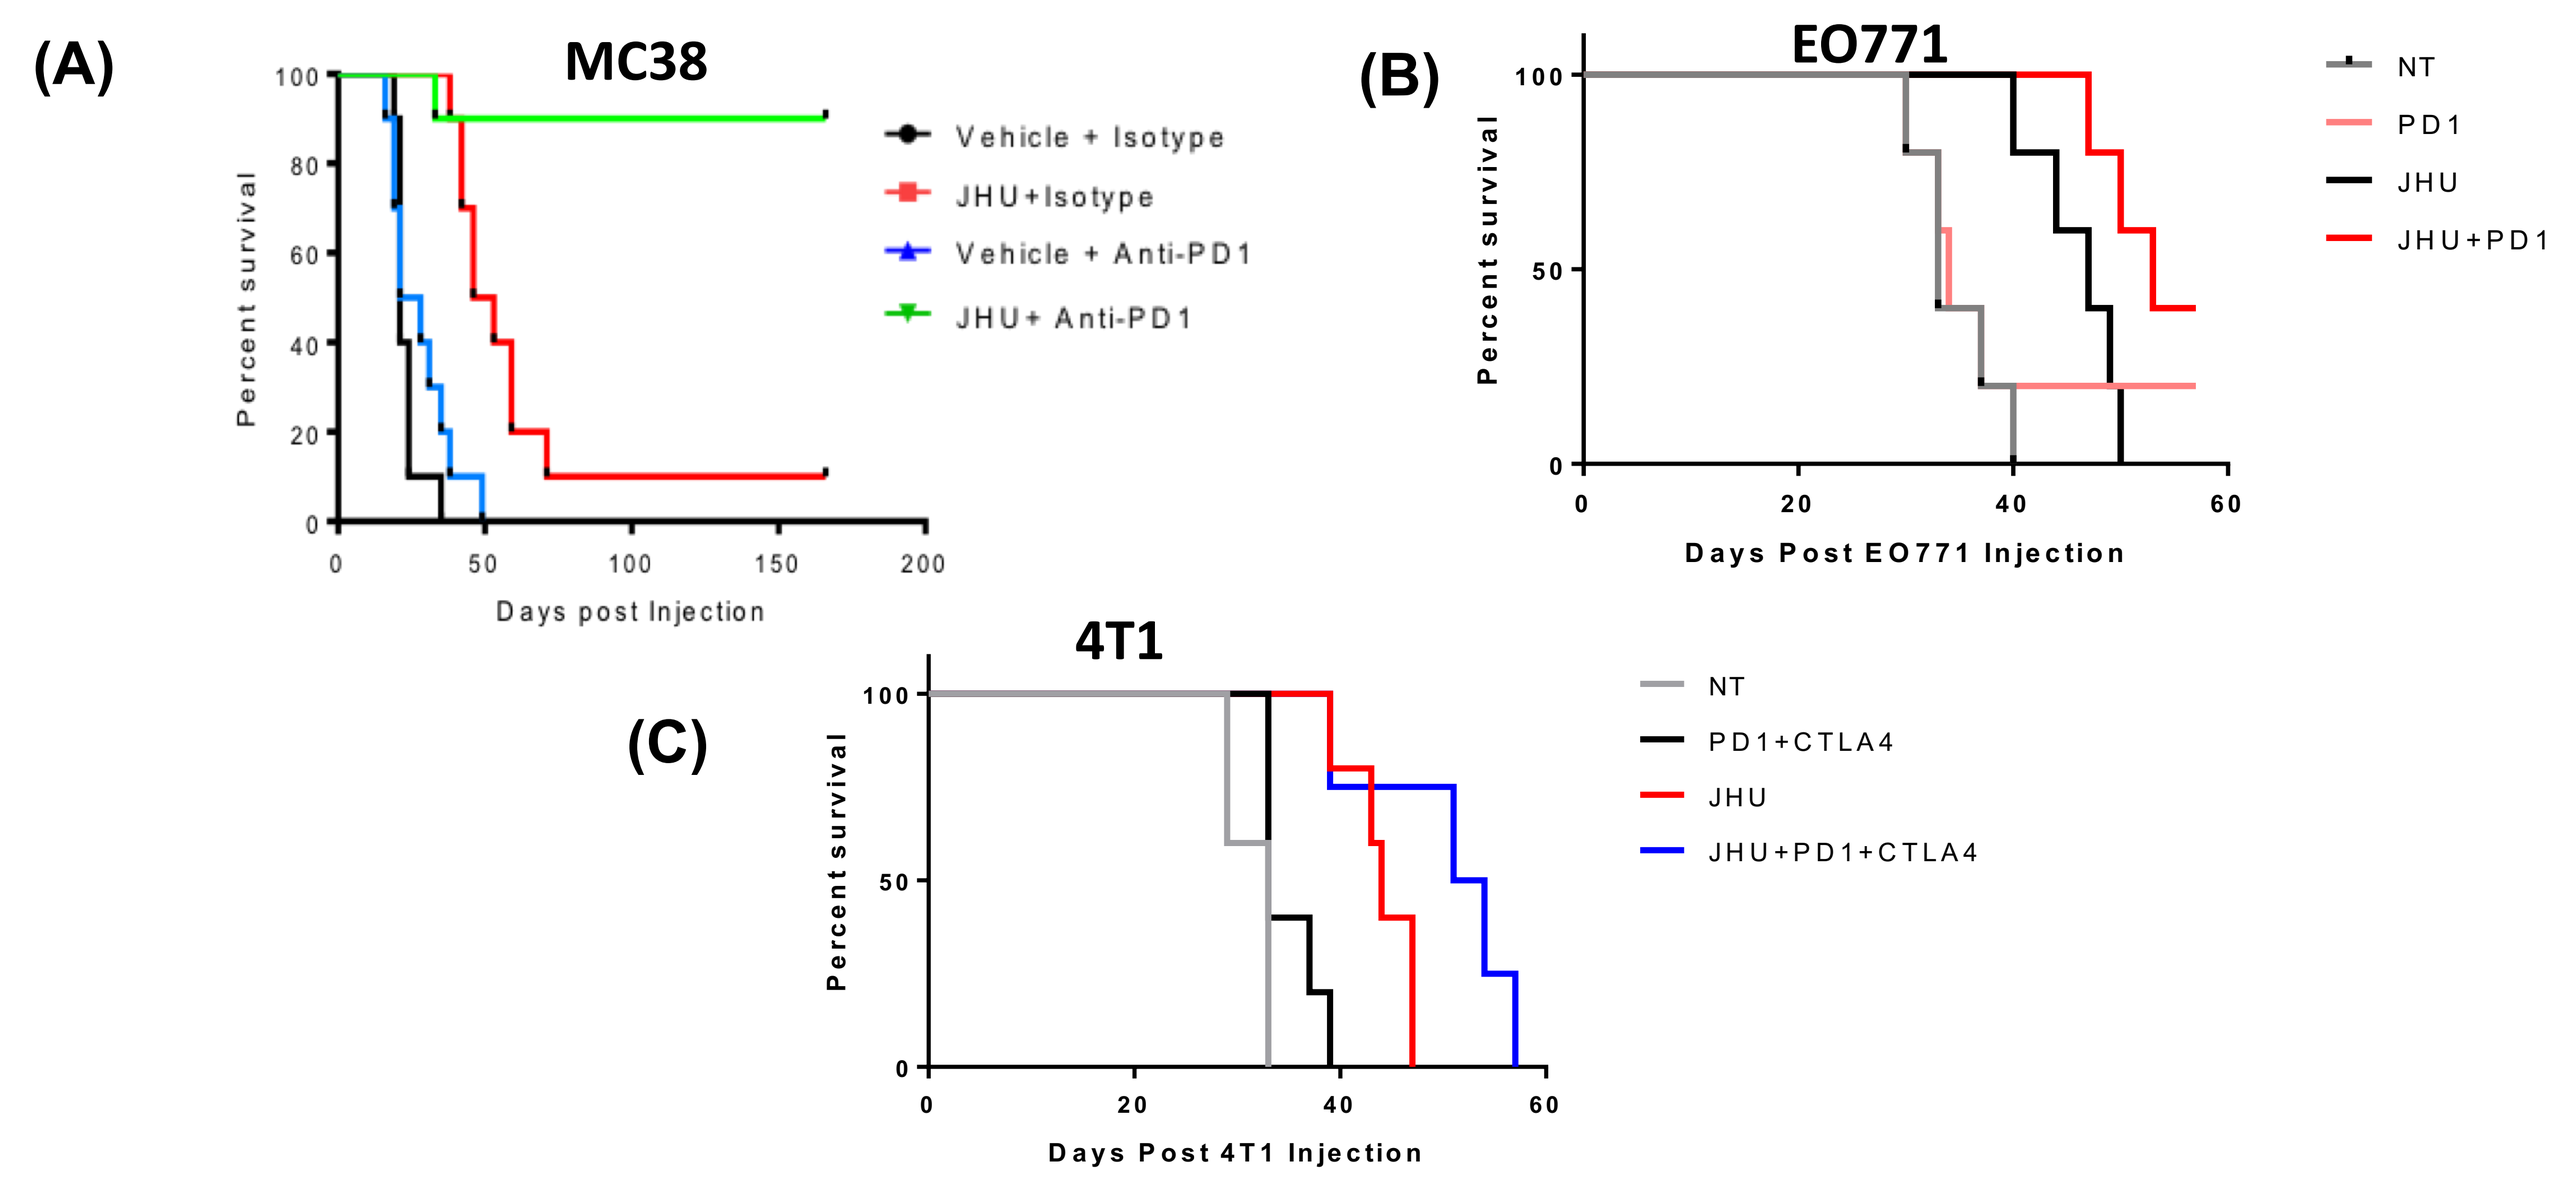
(A)
MC38
Vehicle + Isotype
JHU+Isotype
Vehicle + Anti-PD1
JHU+ Anti-PD1
Percent survival
Days post Injection
(B)
EO771
NT
PD1
JHU
JHU+PD1
Days Post EO771 Injection
4T1
(C)
NT
PD1+CTLA4
JHU
JHU+PD1+CTLA4
Days Post 4T1 Injection

Fig 7. (A) glutamine inhibition enhances (A) aPD1 therapy in MC38 tumor model, susceptible tumors to immune checkpoint blockade (B) aPD1 therapy in EO771 tumor model, mildly susceptible to immune checkpoint blockade (C) aPD1 and aCTLA4 therapy in 4T1 tumor model resistant to immune checkpoint blockade

Summary & Conclusion

Targeting glutamine metabolism using JHU-083 enhances immunotherapy for cancer

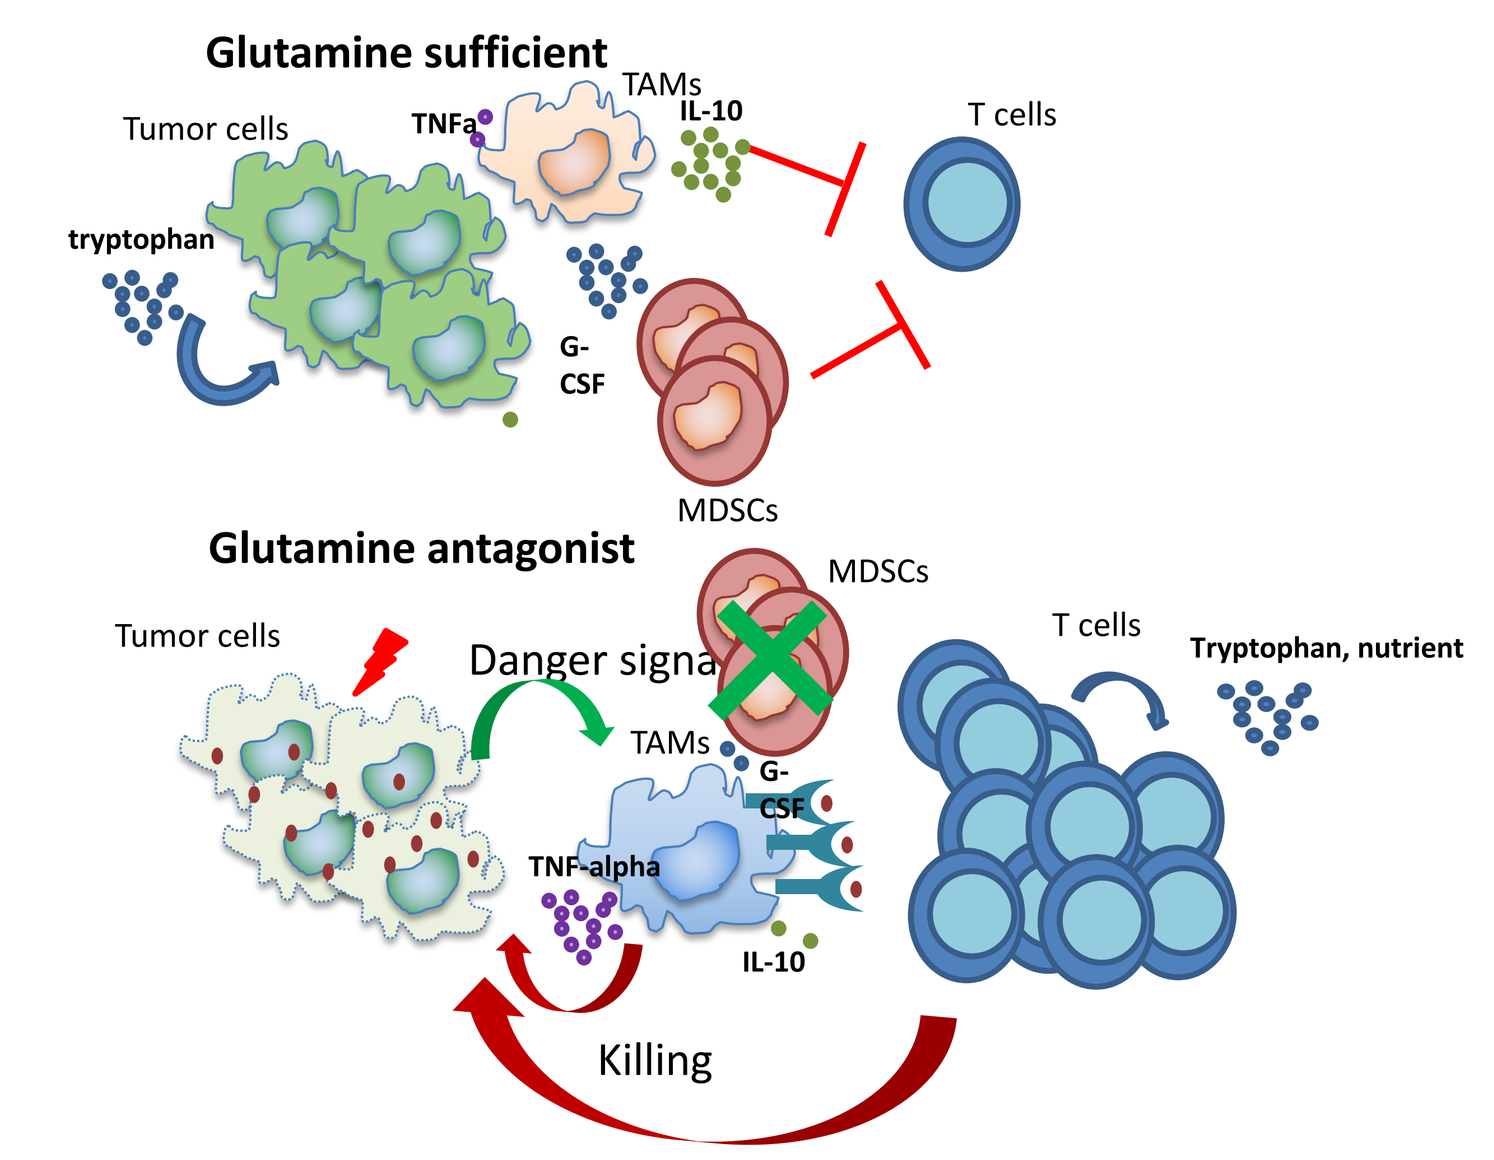
Glutamine sufficient
Tumor cells
TNFa
TAMs
IL-10
T cells
tryptophan
G-CSF
MDSCs
Glutamine antagonist
Tumor cells
Danger signal
MDSCs
T cells
Tryptophan, nutrient
TAMs
TNF-alpha
IL-10
Killing

- **Inhibition of glutamine metabolism leads to reduced MDSCs**
- **In part, this decrease is due to decreased CSF3 secretion from TAMs**
- **Inhibition of glutamine metabolism leads to markedly decreased kynurenine in both primary tumors and metastatic sites, correlating with reduced MDSCs in the lung**
- **Consequently, JHU-083 markedly inhibits lung metastasis**
- **Treatment with JHU-083 enhances inflammatory TAMs**
- **Treatment with JHU-083 leads to enhanced immunologic cell death resulting in enhanced tumor antigen presentation and activation**
- **Treatment with JHU-083 is able to enhance immunotherapy in immune checkpoint blockade resistant tumor**